Safety and efficacy of felzartamab (MOR202) in anti-phospholipase A2 receptor (PLA2R) autoantibody-positive membranous nephropathy (MN): The M-PLACE study

Brad H. Rovin,[1]* Pierre M. Ronco,[2,3] Jack F. M. Wetzels,[4] Sharon G. Adler,[5] Isabelle Ayoub,[1] Philippe Zaoui,[6] Seung Hyeok Han,[7] Julia Jauch-Lembach,[8] Nicola Faulhaber,[8] Jie Cui,[9] Stefan Härtle,[8] Ben Sprangers[10]

[1]The Ohio State University Wexner Medical Center, Columbus, OH, USA; [2]Sorbonne Université and INSERM UMRS 1155, Paris, France; [3]Centre Hospitalier Le Mans, Le Mans, France; [4]Radboud University Medical Center, Nijmegen, The Netherlands; [5]Lundquist Research Institute at Harbor UCLA, Torrance, CA, USA; [6]Centre Hospitalier Universitaire Grenoble Alpes, Grenoble, France; [7]Yonsei University College of Medicine, Seoul, South Korea; [8]MorphoSys AG, Planegg, Germany; [9]MorphoSys US Inc., Boston, MA, USA; [10]University Hospitals Leuven, Leuven, Belgium.

Poster SA-PO679 – Glomerular Diseases: Clinical, Outcomes, Trials – III; November 5, 2022

Background

- Primary MN is a rare autoimmune kidney disease and a leading cause of nephrotic syndrome in adults worldwide[1,2]
- Approximately 75% of primary MN cases arise due to autoantibodies to PLA2R, a cell surface protein expressed on podocytes.[3,4] Anti-PLA2R autoantibodies bind to podocyte PLA2R, forming immune complexes that trigger complement activation and inflammation, impairing glomerular function[1]
- Autoantibodies are secreted by autoreactive CD20-/CD38+ plasma cells and plasmablasts;[5] high anti-PLA2R autoantibody titers are associated with more severe disease, a longer time to disease remission, and a decreased response to anti-CD20 rituximab treatment[3,6,7]
- Treatment options for patients with MN are limited, and the current standard of care comprises off-label conservative therapy and immunosuppressive therapy (IST) when indicated, such as alkylating agents, anti-CD20 antibodies, and calcineurin inhibitors.[1,8] However, current treatment approaches are not effective in all patients and, in particular, conventional ISTs are associated with a substantial risk of toxicity and unsatisfactory remission rates[1,3,8]
- Felzartamab is an investigational, fully human IgG1 monoclonal anti-CD38 antibody that depletes plasmablasts and plasma cells via antibody-dependent cellular cytotoxicity and antibody-dependent cellular phagocytosis[9,10] (**Figure 1**)
 - Its proposed mechanism of action is independent of complement-dependent cytotoxicity, which has been associated with infusion-related reactions (IRRs)

Figure 1. Proposed mechanism of action of felzartamab in MN

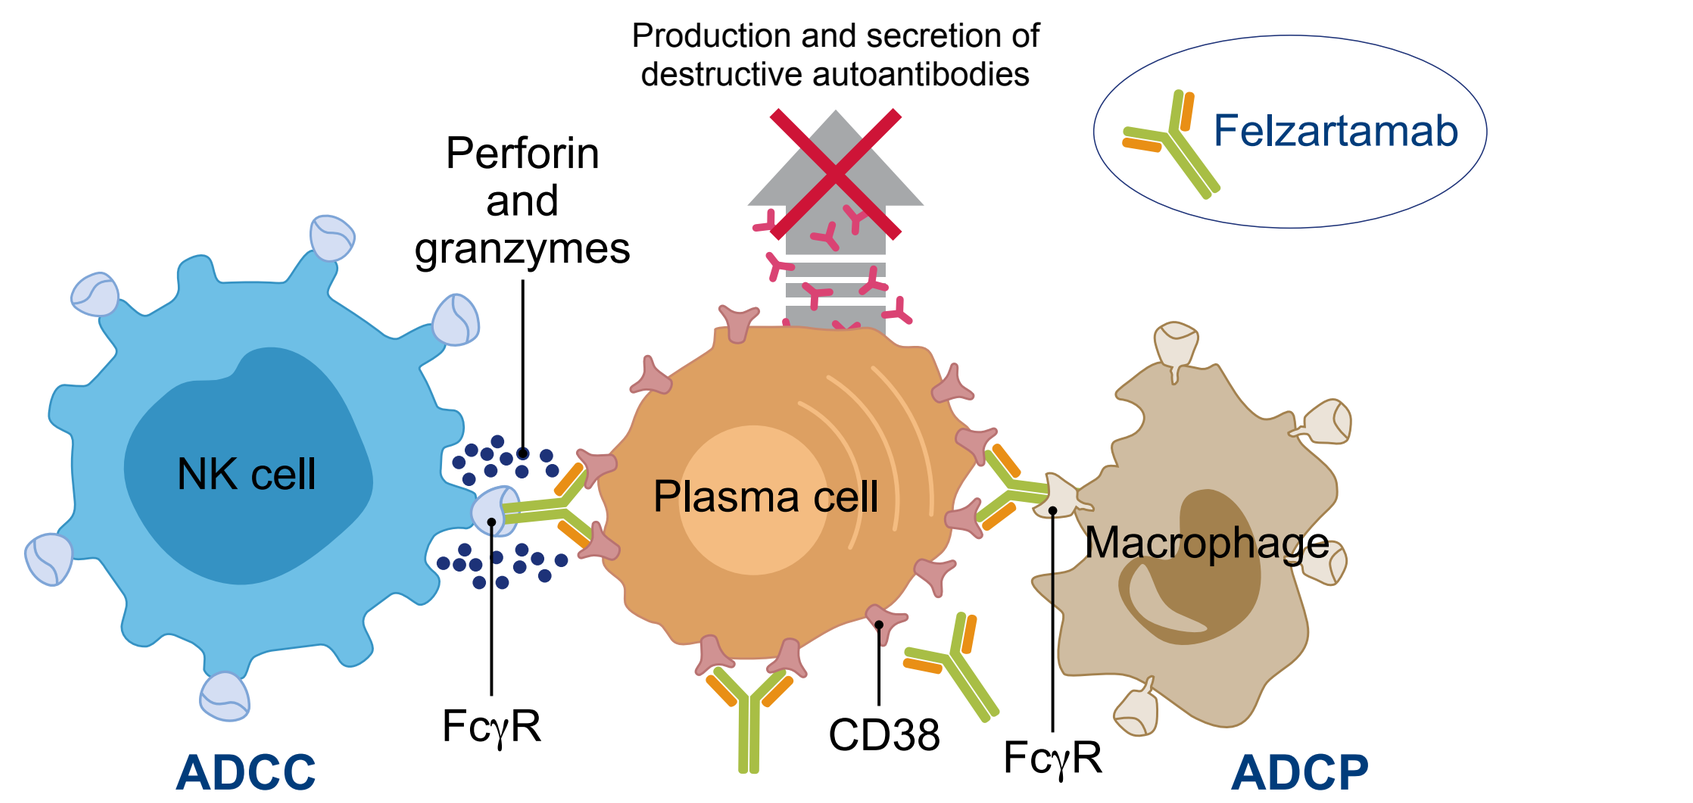
Production and secretion of destructive autoantibodies
Felzartamab
Perforin and granzymes
NK cell
Plasma cell
Macrophage
FcγR
CD38
FcγR
ADCC
ADCP

Plasmablasts and plasma cells are CD38+ cells.
ADCC, antibody-dependent cellular cytotoxicity; ADCP, antibody-dependent cellular phagocytosis; CD, cluster of differentiation; FcγR, Fc-gamma receptor; NK, natural killer.

- M-PLACE (NCT04145440) is a Phase Ib/IIa, proof of concept, open-label, multicenter, multinational study to assess the safety and efficacy of felzartamab in adults with anti-PLA2R antibody-positive MN
- A preliminary analysis showed the safety profile of felzartamab is consistent with its proposed mechanism of action, and the potential of felzartamab to deplete plasma cells is supported by the reduction of anti-PLA2R antibody titers and urine protein:creatinine ratio (UPCR)[11]
- Patients with MN in this study have been shown to mount humoral responses to COVID-19 vaccination following felzartamab treatment, consistent with the selectivity of felzartamab to reduce CD20-/CD38+ plasma cells whilst leaving the early B-cell compartment intact[12]
- Here we present an updated analysis of safety and efficacy from the ongoing M-PLACE study with a longer patient follow-up (data cut-off: April 10, 2022)

Patients and methods

- Enrolled patients were aged 18–80 years with biopsy-proven anti-PLA2R antibody-positive MN requiring IST according to the site investigator. Other key inclusion criteria were:
 - UPCR ≥3.0 g/g or proteinuria ≥3.5 g/24 h from a 24-hour urine screening
 - Estimated glomerular filtration rate (eGFR) ≥50 mL/min/1.73m², or eGFR ≥30 and <50 mL/min/1.73 m² with interstitial fibrosis and tubular atrophy score of <25% in a kidney biopsy
- Patients were enrolled into one of two cohorts:
 - **Cohort 1:** newly diagnosed patients and those who relapsed after a prior therapy; patients had serum anti-PLA2R antibodies ≥50 RU/mL at screening (central laboratory, Euroimmun ELISA)
 - **Cohort 2:** patients refractory to a prior IST, i.e. patients who did not achieve immunologic remission after prior IST(s) and are without promising standard therapeutic options; patients had serum anti-PLA2R antibodies ≥20 RU/mL measured at screening (central laboratory, Euroimmun ELISA)
- Patients were planned to receive nine doses of felzartamab 16 mg/kg intravenously, once weekly during the first 4 weeks and once every 4 weeks thereafter for a 24-week treatment (**Figure 2**). After the end of treatment (EOT), patients remained in follow-up for up to 28 weeks

Figure 2. Study design

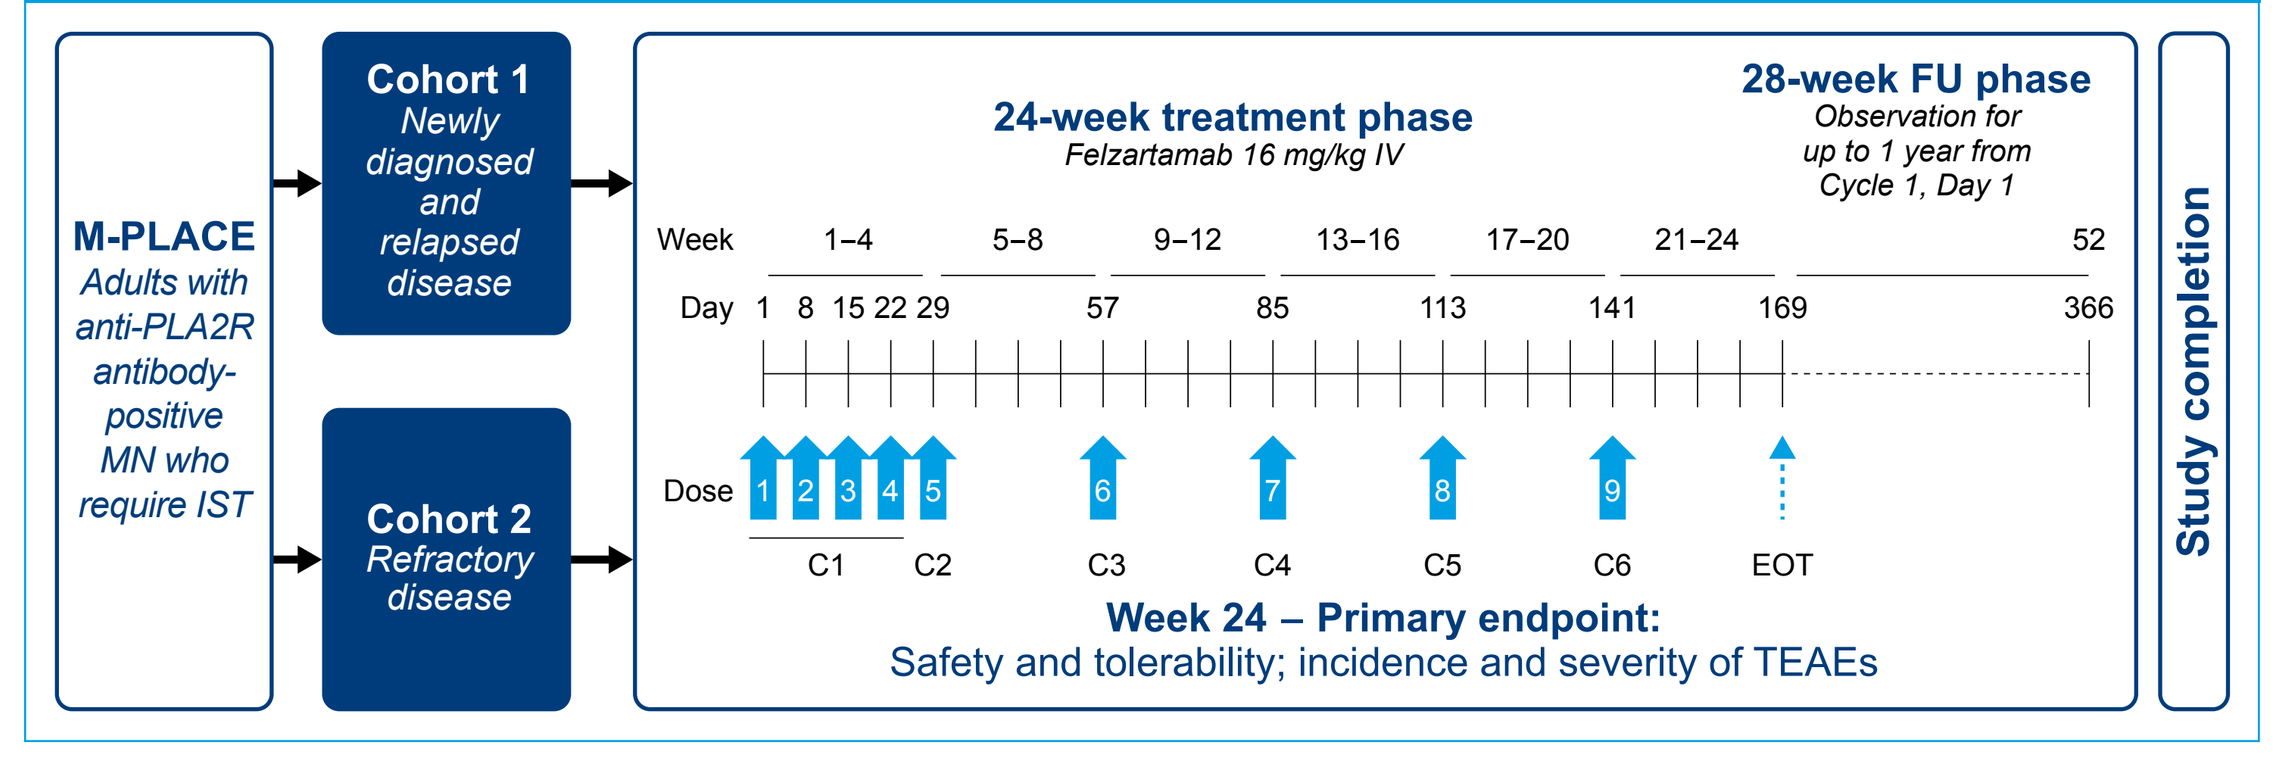
M-PLACE
Adults with anti-PLA2R antibody-positive MN who require IST
Cohort 1
Newly diagnosed and relapsed disease
Cohort 2
Refractory disease
24-week treatment phase
Felzartamab 16 mg/kg IV
28-week FU phase
Observation for up to 1 year from Cycle 1, Day 1
Week 1–4 5–8 9–12 13–16 17–20 21–24 52
Day 1 8 15 22 29 57 85 113 141 169 366
Dose 1 2 3 4 5 6 7 8 9
C1 C2 C3 C4 C5 C6 EOT
Week 24 – Primary endpoint:
Safety and tolerability; incidence and severity of TEAEs
Study completion

Cohort 1: serum anti-PLA2R antibodies ≥50 RU/mL; Cohort 2: serum anti-PLA2R antibodies ≥20 RU/mL.
C, cycle; EOT, end of treatment; FU, follow-up; IST, immunosuppressive treatment; IV, intravenous; MN, membranous nephropathy; PLA2R, phospholipase A2 receptor; TEAE, treatment-emergent adverse event.

- The primary endpoint was the incidence and severity of treatment-emergent adverse events (TEAEs)
 - TEAEs were graded according to the National Cancer Institute Common Terminology Criteria for Adverse Events, version 5.0
- Key secondary endpoints were best immunologic response, rate of immunologic complete response (ICR), and immunologic partial response (IPR) based on reduction of serum anti-PLA2R antibody titer
 - ICR was defined as a reduction in anti-PLA2R antibody titer to <14 RU/mL
 - IPR was defined as a ≥50% reduction in anti-PLA2R antibody titer from baseline
- Exploratory endpoints included the effect of treatment on reduction of proteinuria, as expressed by proteinuria response at the EOT phase (end of Cycle 6) and end of study (EOS) (after 12 months), renal function (change in eGFR from baseline at EOT and EOS), and change of serum albumin levels from baseline at EOT and EOS
- All statistical analyses use descriptive summary statistics. Study is ongoing; no formal database lock was performed, therefore data may be subject to change

Results

Demographics and baseline characteristics

- At the data cut-off, 31 patients had been enrolled (Cohort 1: 18 patients; Cohort 2: 13 patients) from 21 sites in North America (5), Asia-Pacific (3), and Europe (13), of whom 23 patients completed the treatment phase (**Figure 3**)
- The mean age of enrolled patients was 57.5 years, 48.4% received one or more ISTs, the median baseline circulating anti-PLA2R antibody titer was 161.0 U/mL, the median baseline UPCR was 6.1 g/g, the median baseline eGFR was 61.0 mL/min/1.73m², and the median baseline serum albumin was 2.6 g/dL **(Table 1)**

Figure 3. Patient disposition

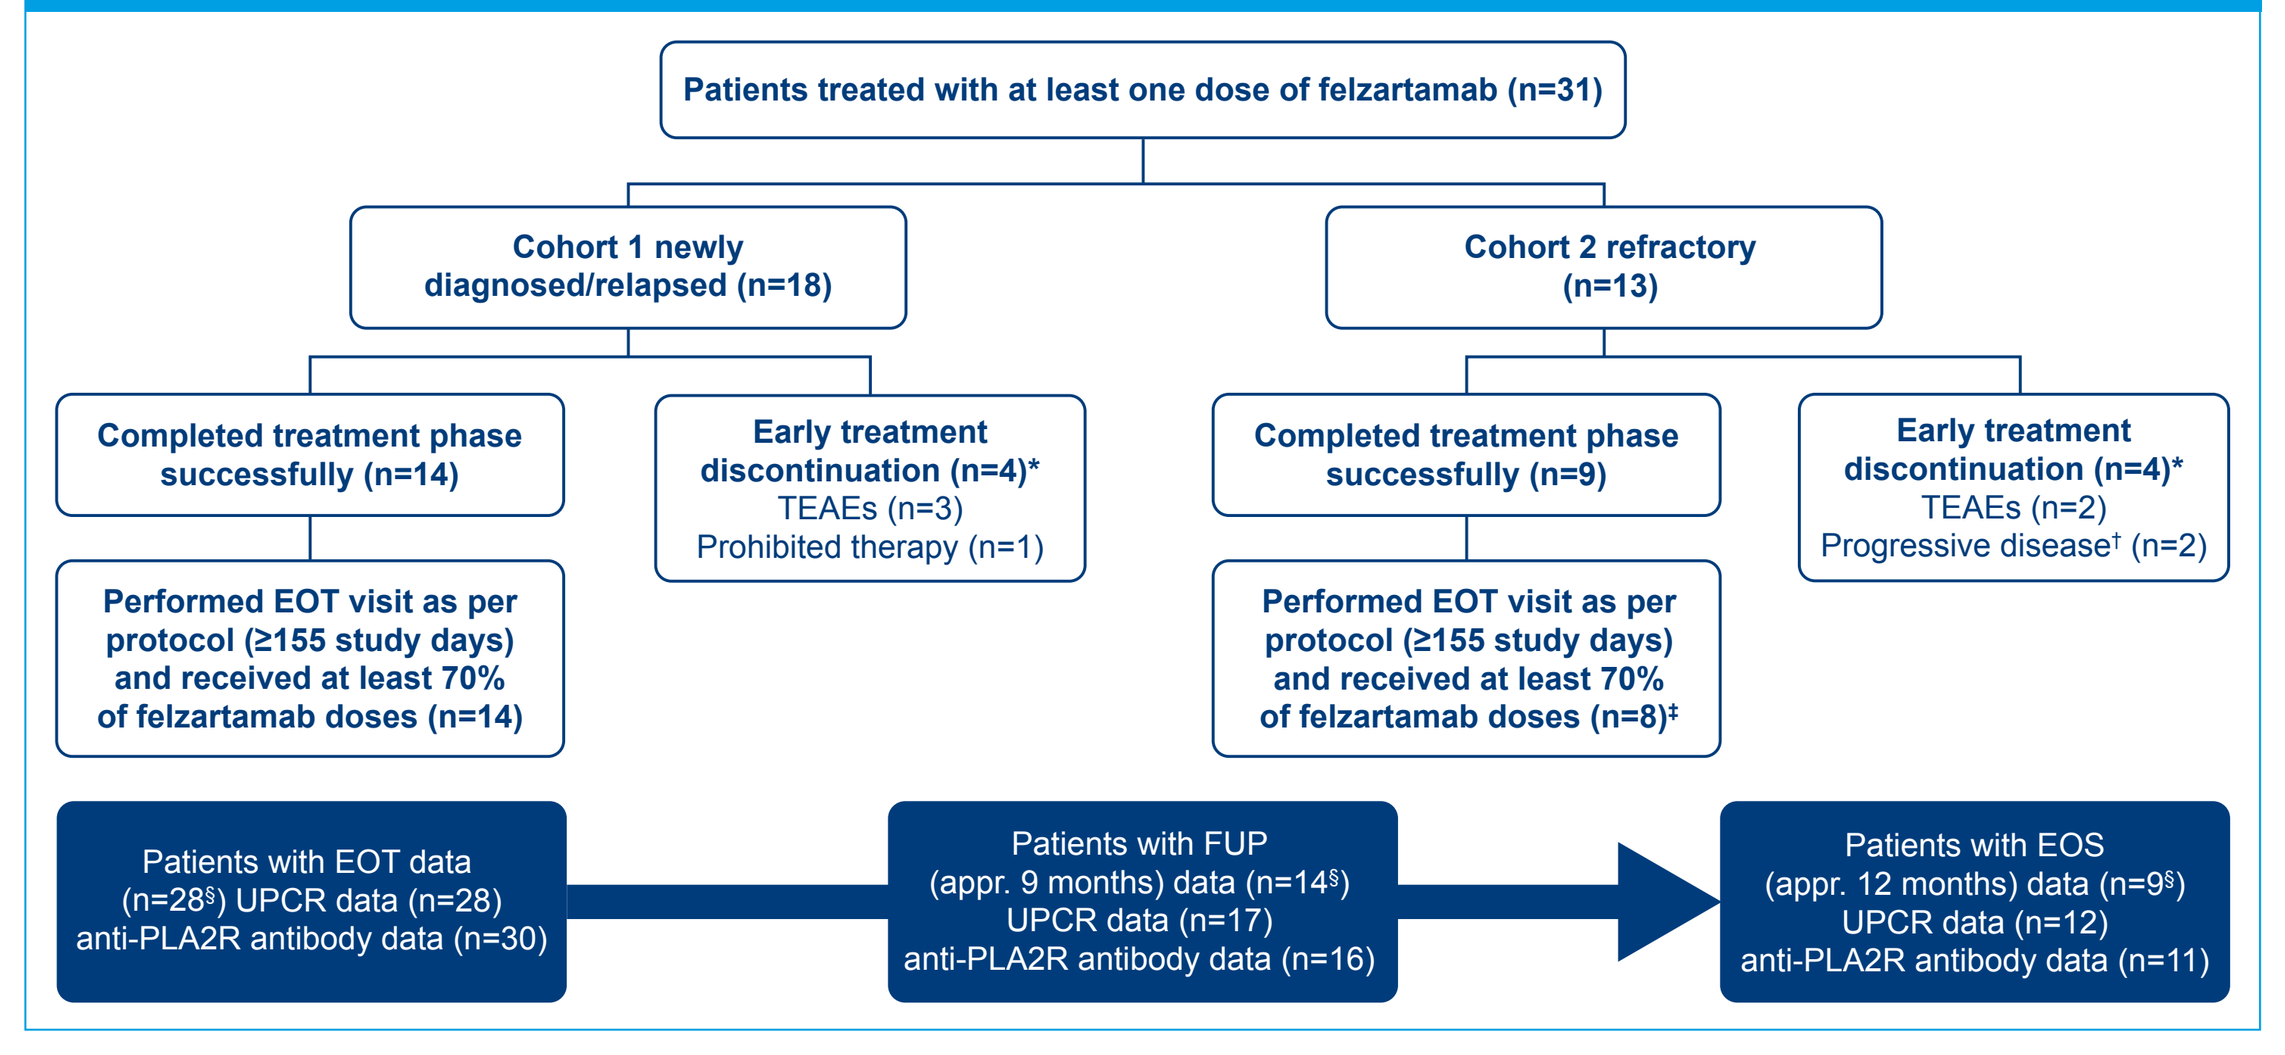
Patients treated with at least one dose of felzartamab (n=31)
Cohort 1 newly diagnosed/relapsed (n=18)
Cohort 2 refractory (n=13)
Completed treatment phase successfully (n=14)
Early treatment discontinuation (n=4)*
TEAEs (n=3)
Prohibited therapy (n=1)
Completed treatment phase successfully (n=9)
Early treatment discontinuation (n=4)*
TEAEs (n=2)
Progressive disease† (n=2)
Performed EOT visit as per protocol (≥155 study days) and received at least 70% of felzartamab doses (n=14)
Performed EOT visit as per protocol (≥155 study days) and received at least 70% of felzartamab doses (n=8)‡
Patients with EOT data (n=28§) UPCR data (n=28) anti-PLA2R antibody data (n=30)
Patients with FUP (appr. 9 months) data (n=14§) UPCR data (n=17) anti-PLA2R antibody data (n=16)
Patients with EOS (appr. 12 months) data (n=9§) UPCR data (n=12) anti-PLA2R antibody data (n=11)

*Of the eight patients who discontinued treatment early, six patients discontinued the study. In total, seven patients discontinued the study (n=6 for use of prohibited therapy; n=1 due to Principal Investigator's decision). †Defined as a >30% decrease of eGFR from baseline, or increase in UPCR determined from 24 h urine collection by >50% from baseline value and <10% decline of anti-PLA2R antibody titers compared to baseline. ‡One patient had missing evaluation. §Patients with both UPCR and anti-PLA2R antibody data.
eGFR, estimated glomerular filtration rate; EOS, end of study; EOT, end of treatment; FUP, follow-up phase; IRR, infusion-related reaction; PLA2R, phospholipase A2 receptor; TEAE, treatment-emergent adverse event; UPCR, urine protein:creatinine ratio.

Table 1. Baseline characteristics

Characteristic	Cohort 1 (n=18)	Cohort 2 (n=13)	All patients (N=31)
Gender, n (%)			
Male	13 (72.2)	11 (84.6)	24 (77.4)
Female	5 (27.8)	2 (15.4)	7 (22.6)
Age (years) at screening, mean (SD)	59.2 (11.3)	55.0 (12.5)	57.5 (11.8)
Prior IST,* n (%)			
None	16 (88.9)	0	16 (51.6)
1	0	7 (53.8)	7 (22.6)
2	1 (5.6)	2 (15.4)	3 (9.7)
≥3	1 (5.6)	4 (30.8)	5 (16.1)
Baseline anti-PLA2R* antibody titer (RU/mL),† median (range)	182.0 (67.0–514.0)	122.0 (10.0–1,075.0)	161.0 (10.0–1,075.0)
Baseline UPCR (g/g),‡ median (range)	5.9 (3.1–10.5)	6.2 (3.5–10.6)	6.1 (3.1–10.6)
Baseline eGFR (mL/min/1.73m²), median (range)	64.5 (34.0–114.0)	60.0 (29.0–82.0)	61.0 (29.0–114.0)
Baseline serum albumin (g/dL),§ median (range)	2.5 (1.9–3.7)	2.8 (2.2–3.8)	2.6 (1.9–3.8)

*Prior to entering the trial, two patients in Cohort 1 and six patients in Cohort 2 were treated with cyclophosphamide for anti-PLA2R antibody-positive MN. †Baseline anti-PLA2R antibody titer is the last value prior to treatment start. ‡Baseline UPCR value is defined as the mean of the values determined at screening and prior to first dose on Cycle 1 Day 1. §Reference range of albumin in central labs: 3.5–5.2 g/dL.
eGFR, estimated glomerular filtration rate; IST, immunosuppressive treatment; MN, membranous nephropathy; PLA2R, phospholipase A2 receptor; SD, standard deviation; UPCR, urine protein:creatinine ratio.

Safety

- The safety profile was consistent with the proposed mechanism of action of felzartamab, and TEAEs were manageable (**Table 2**)
- Overall, 27 (87%) patients had at least one TEAE, which were mostly mild/moderate in severity and the majority resolved; no event had a fatal outcome
- The most frequently reported TEAE was IRR (29% of patients; one Grade 3)
 - Most IRRs resolved within 1 or 2 days of onset
 - One patient experienced a serious Grade 3 IRR with symptoms of hoarseness, coughing, dyspnea, fatigue, weakness, hypertension, tachycardia approximately 30 minutes after start of infusion; felzartamab was permanently discontinued and the event resolved within 2 days of its onset
 - All IRRs were reported during the first infusion, except one event that was reported during the third infusion

Table 2. Summary of most frequent TEAEs regardless of relationship (≥2 patients of overall patient population)

	Overall (N=31)	Cohort 1 (n=18)	Cohort 2 (n=13)
Adverse reaction SOC/PT, n (%)	Any grade	CTCAE Grade ≥3	
Blood and lymphatic system disorders			
Neutropenia	3 (9.7)	1 (5.6)	1 (7.7)
Anemia	2 (6.5)	0	0
Gastrointestinal disorders			
Diarrhea	2 (6.5)	0	0
Nausea	5 (16.1)	0	0
General disorders and administration site condition			
Chest pain	3 (9.7)	0	1 (7.7)
Fatigue	3 (9.7)	0	0
Edema peripheral	6 (19.4)	0	2 (15.4)
Pyrexia	2 (6.5)	0	0
Immune system disorders			
Hypogammaglobulinemia	8 (25.8)	0	0
Infection and infestations			
Bronchitis	4 (12.9)	0	0
COVID-19	3 (9.7)	1 (5.6)	0
Injury poisoning and procedural complications			
Infusion-related reaction	9 (29.0)	1 (5.6)	0
Investigations			
Lipase increased	3 (9.7)	0	0
Lymphocyte percentage decreased	2 (6.5)	0	0
Metabolism and nutrition disorders			
Hyperkalemia	2 (6.5)	0	0
Musculoskeletal and connective tissue disorders			
Arthralgia	3 (9.7)	0	0
Back Pain	3 (9.7)	0	0
Myalgia	2 (6.5)	0	0
Rhabdomyolysis	2 (6.5)	0	0
Nervous system disorders			
Headache	3 (9.7)	0	0
Psychiatric disorders			
Insomnia	2 (6.5)	0	0
Respiratory, thoracic and mediastinal disorders			
Cough	2 (6.5)	0	0
Dyspnea	3 (9.7)	0	1 (7.7)

COVID-19, coronavirus disease-2019; CTCAE, Common Terminology Criteria for Adverse Events; PT, preferred term; SOC; System Organ Class; TEAE, treatment-emergent adverse event.

- Five patients (16%) experienced treatment-emergent serious adverse events, three of whom were considered related to felzartamab (Grade 3 type-I hypersensitivity, a Grade 3 IRR, and a Grade 4 neutropenia)
 - Both the type-I hypersensitivity and IRR events resolved within 1 day, and the neutropenia event resolved within 35 days
- Reasons for treatment discontinuations included TEAEs, prohibited therapy, and progressive disease (**Figure 3**)

Immunologic responses

- The majority of patients with available anti-PLA2R antibody data (25/28 [89.3%]) experienced a rapid reduction in anti-PLA2R antibody titers after 1 week of treatment (percent change: median 44.8), regardless of cohort (**Figure 4**)

Figure 4. Relative change from baseline in anti-PLA2R antibody titers at 1 week after the first felzartamab dose (C1D8)

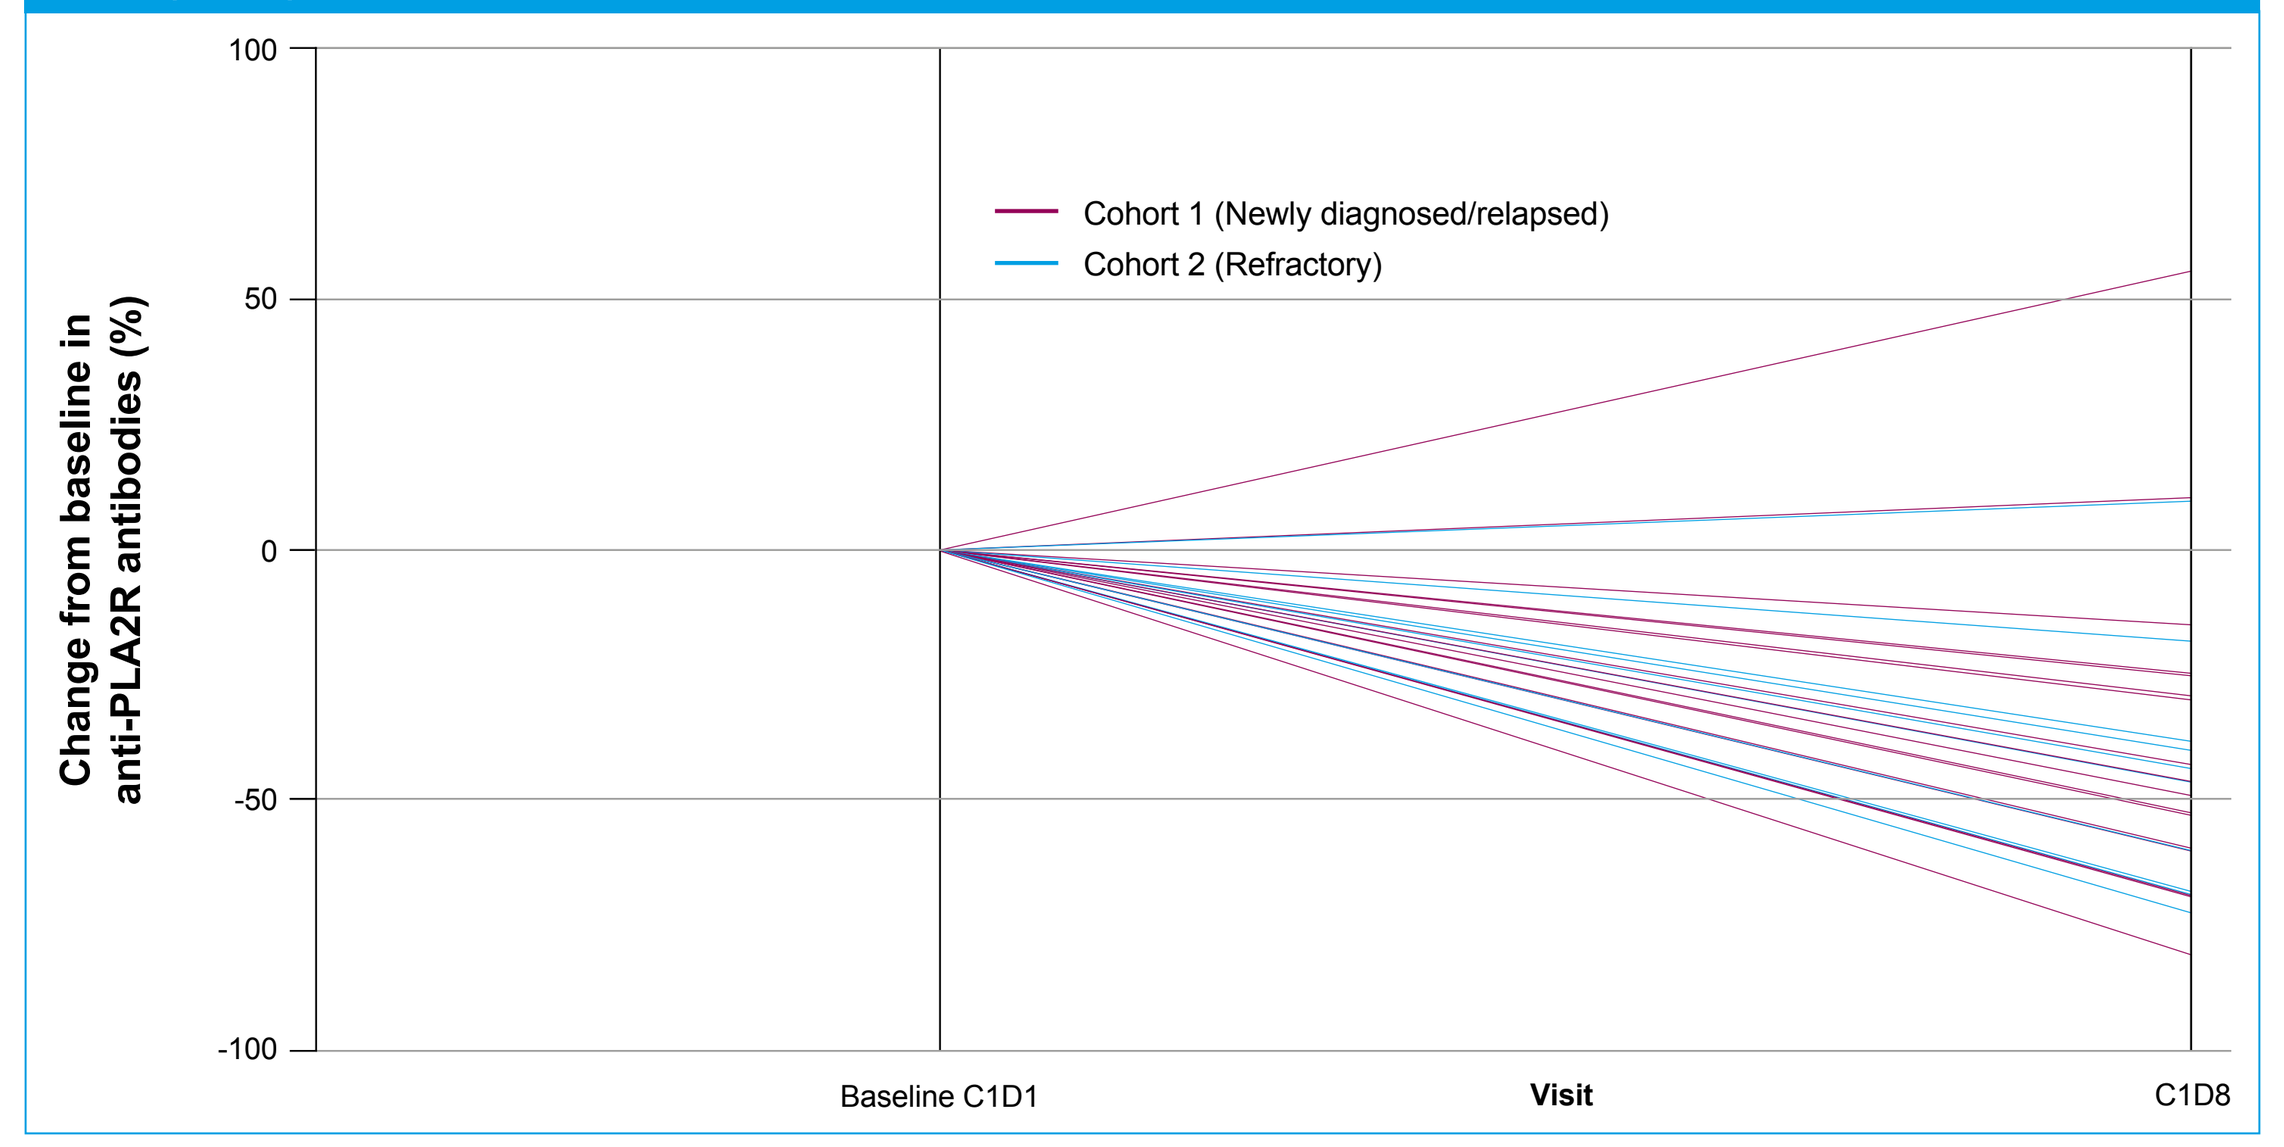
Change from baseline in anti-PLA2R antibodies (%)
100
50
0
-50
-100
Cohort 1 (Newly diagnosed/relapsed)
Cohort 2 (Refractory)
Baseline C1D1
Visit
C1D8

C, cycle; D, day; PLA2R, phospholipase A2 receptor.

- In patients who completed treatment (those who performed their EOT visit as per protocol and who received at least 70% of study treatment: Cohort 1, n=14; Cohort 2, n=8), nine (64.3%) patients in Cohort 1 and three (37.5%) patients in Cohort 2 achieved an IPR (**Figure 5**)
 - Overall, 12 (54.5%) patients achieved an IPR and six (27.3%) patients achieved an ICR at EOT
- For those patients who achieved an IPR at EOT (6 months), the effect was durable in most patients up to Month 12 (end of follow-up period) (**Figure 6**)

Figure 5. Percentage change from baseline in anti-PLA2R titers at EOT (6 months) for patients completing treatment

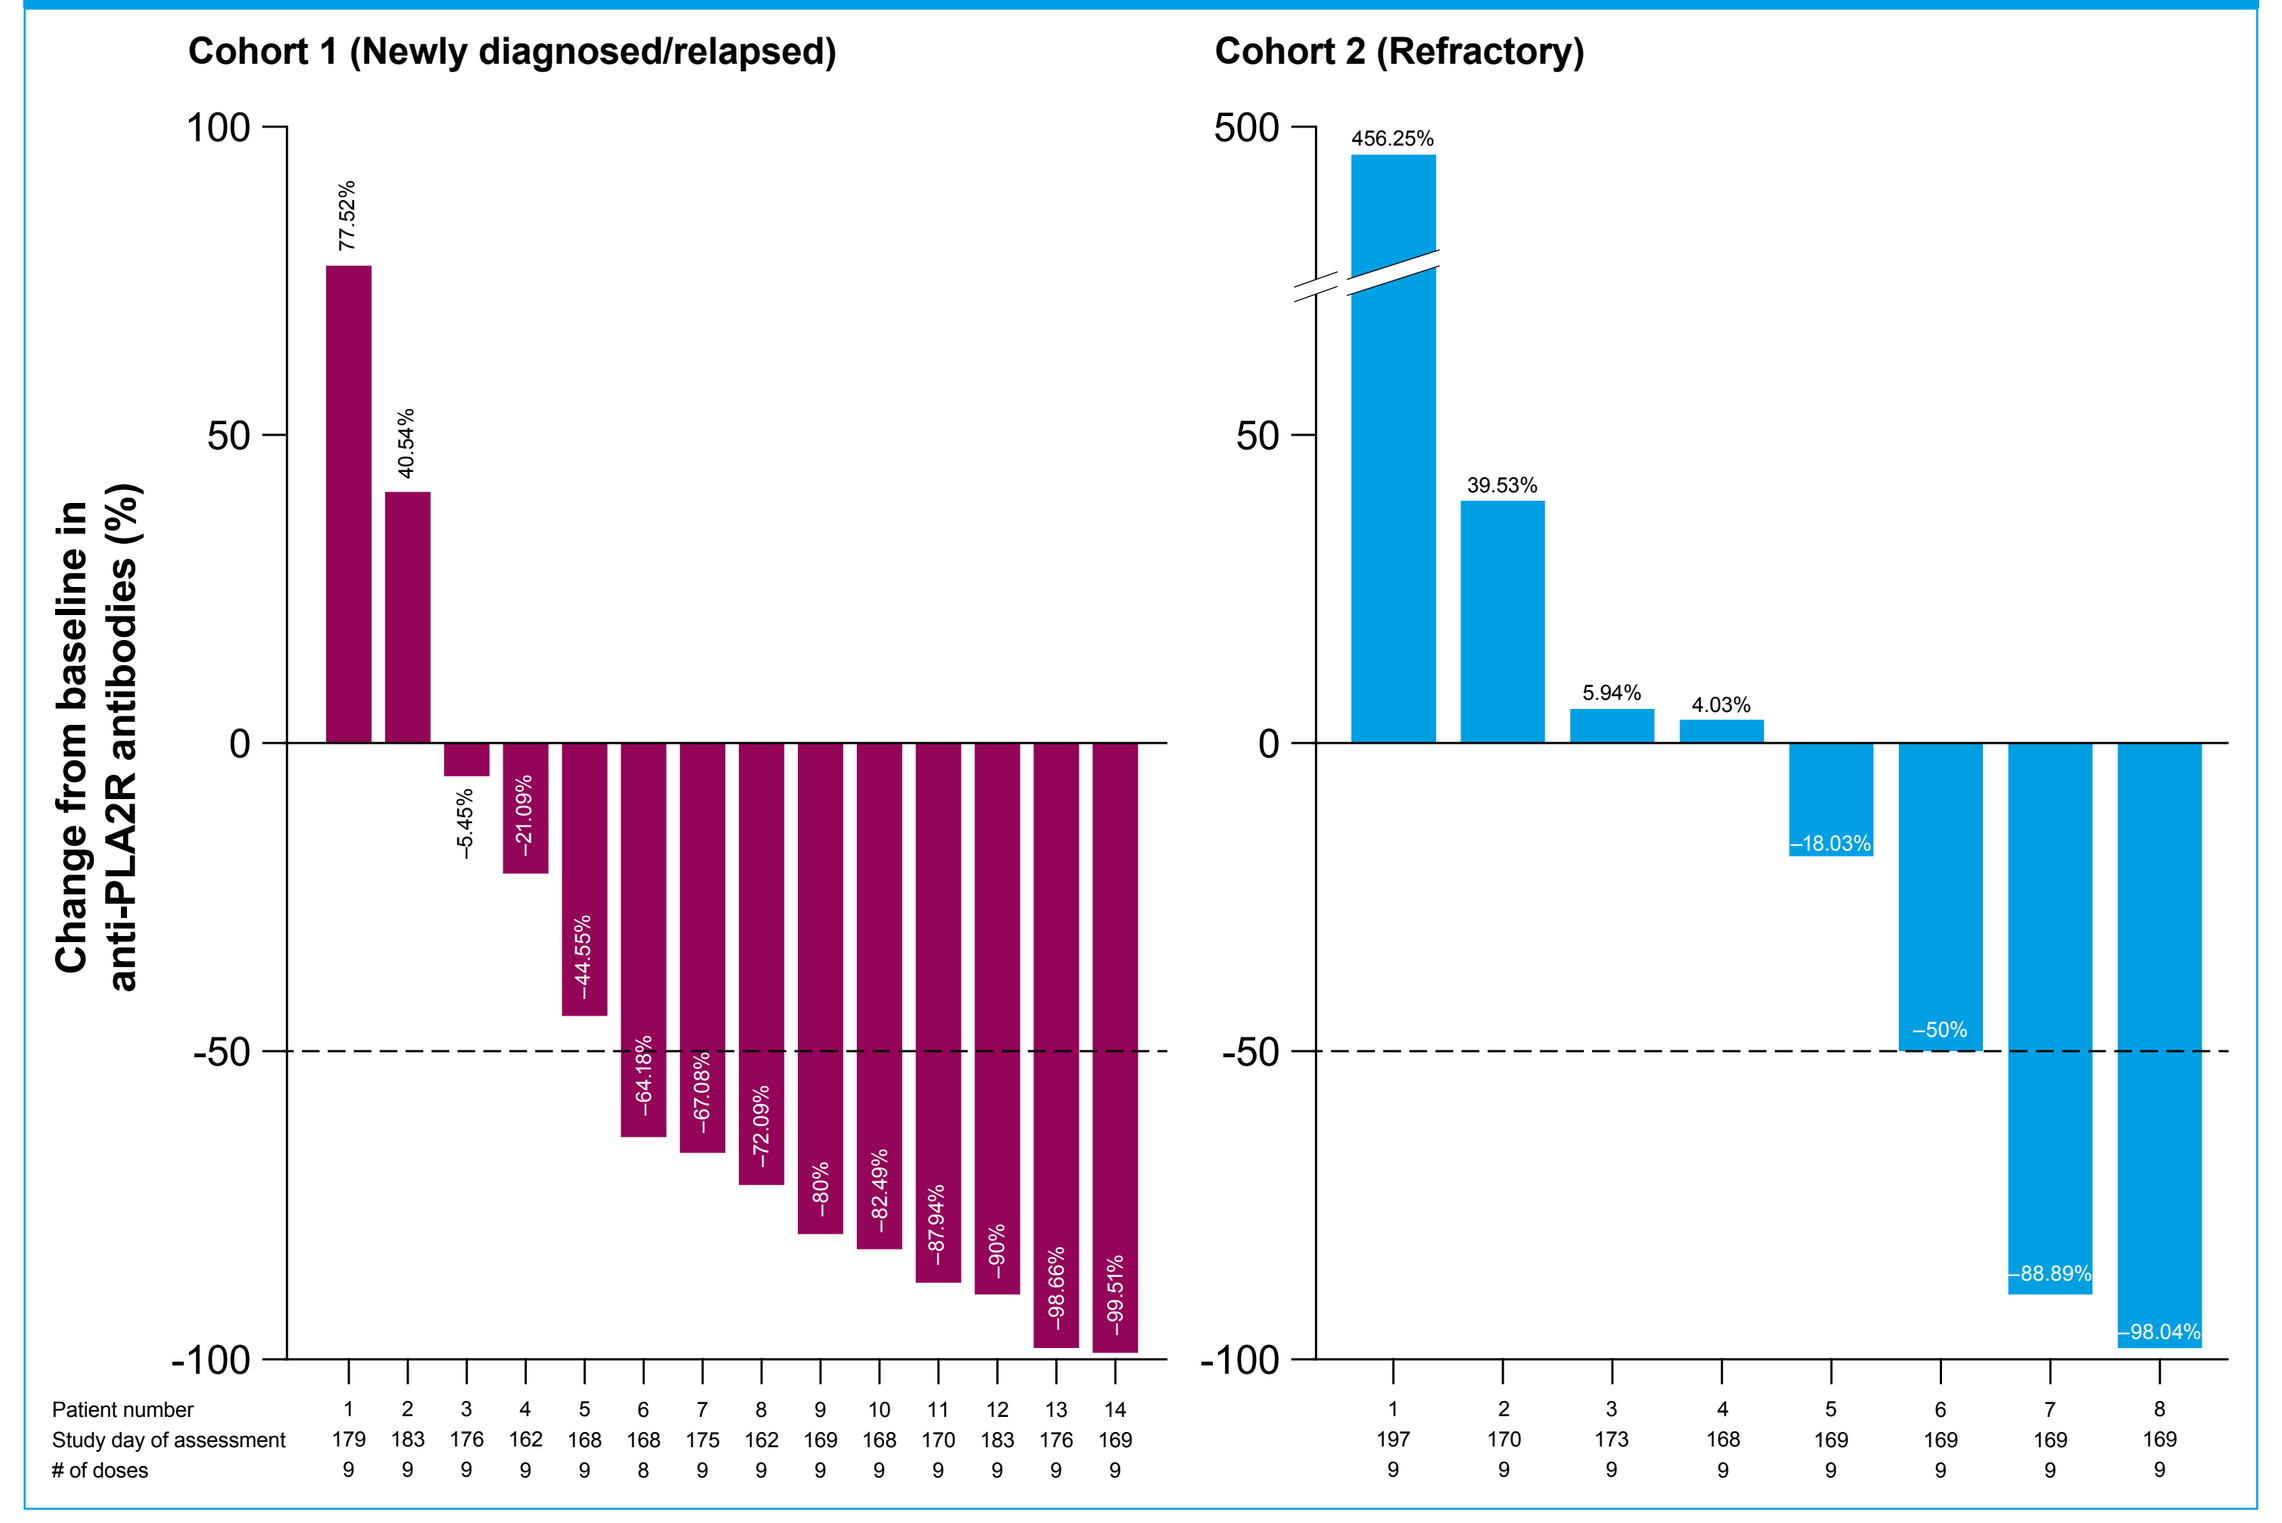
Cohort 1 (Newly diagnosed/relapsed)
Cohort 2 (Refractory)
Change from baseline in anti-PLA2R antibodies (%)
77.52%
40.54%
−5.45%
−21.09%
−44.55%
−64.18%
−67.08%
−72.09%
−80%
−82.49%
−87.94%
−90%
−98.66%
−99.51%
456.25%
39.53%
5.94%
4.03%
−18.03%
−50%
−88.89%
−98.04%
Patient number
Study day of assessment
of doses

EOT, end of treatment; PLA2R, phospholipase A2 receptor.

Figure 6. Percentage change from baseline in anti-PLA2R antibody titer responses in patients with ≥50% response at EOT (6 months) and with available anti-PLA2R antibody titer data post EOT

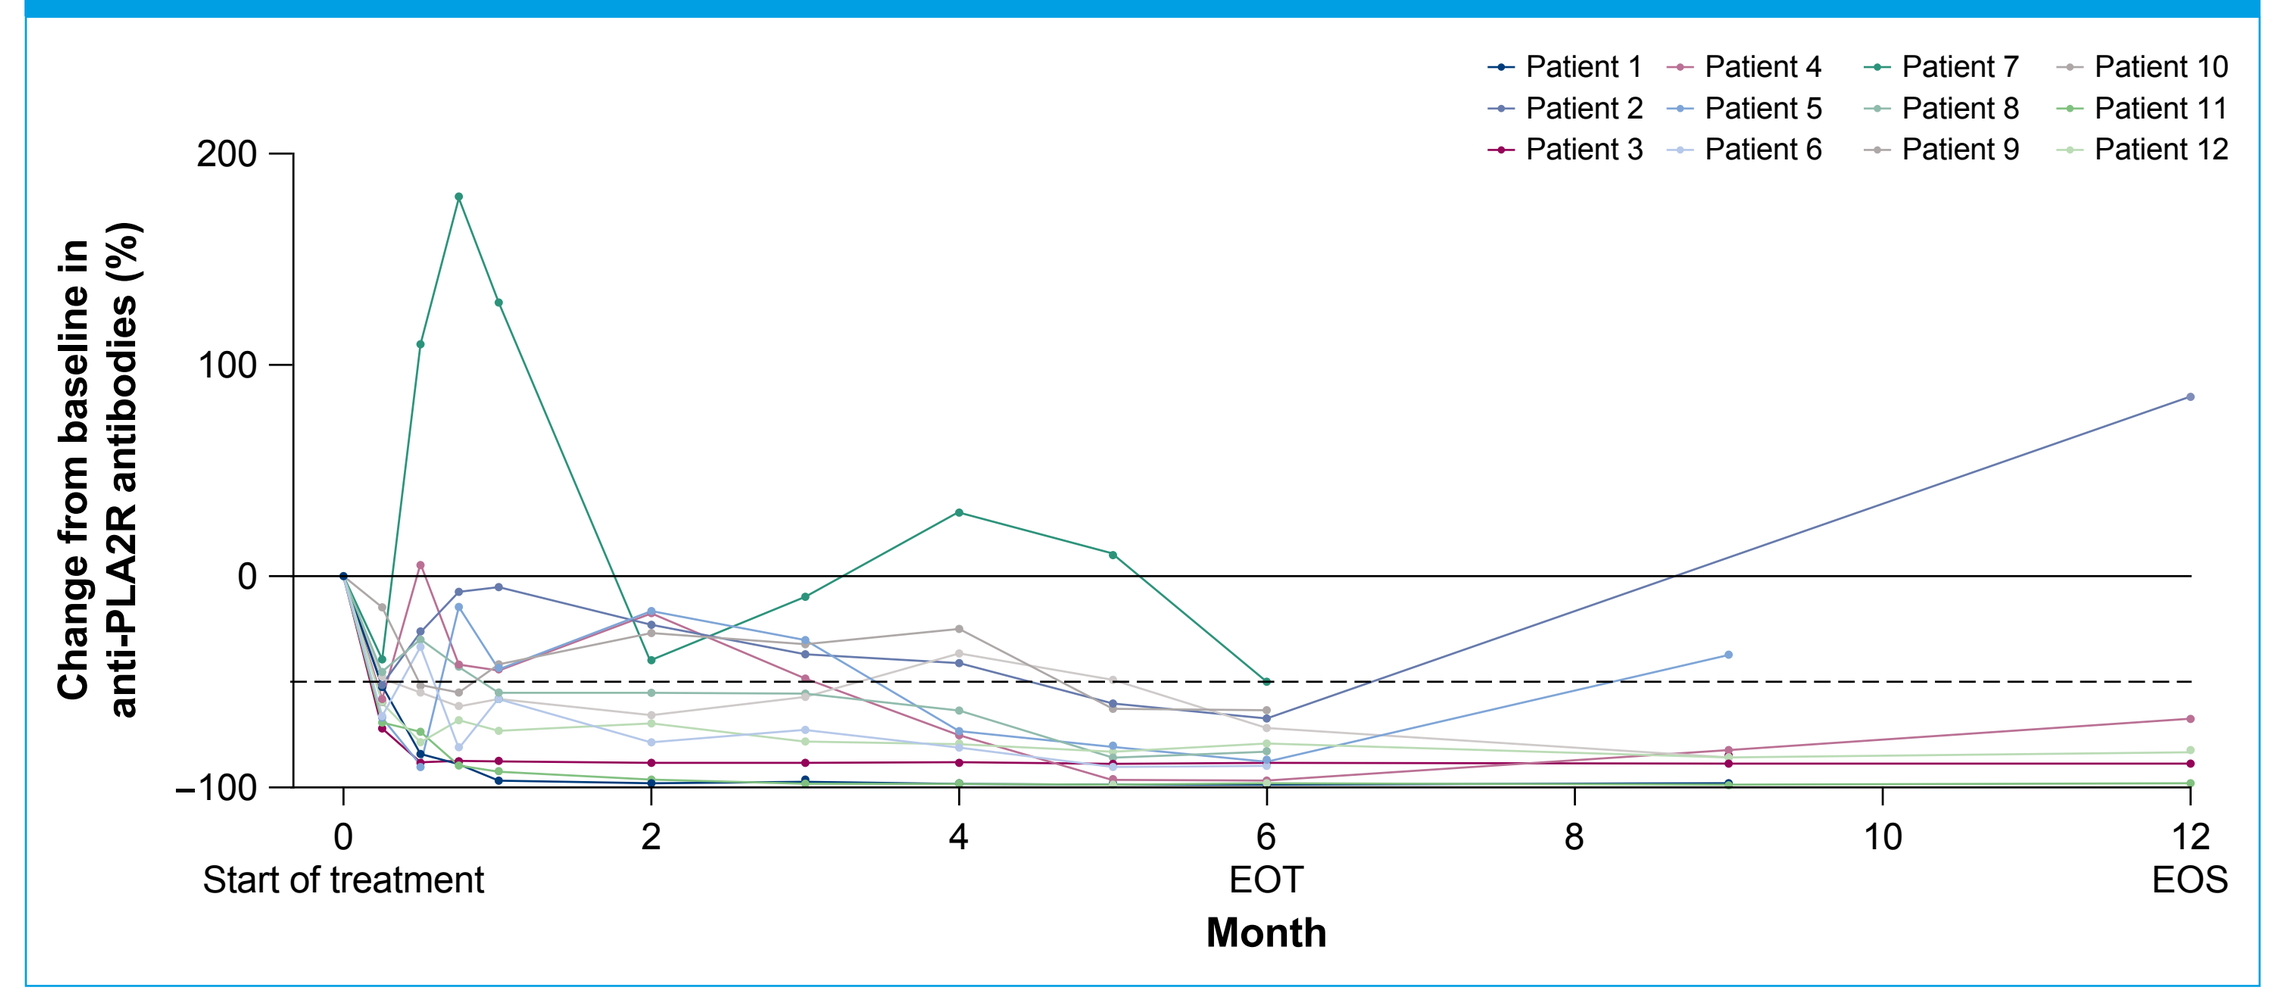
Patient 1
Patient 2
Patient 3
Patient 4
Patient 5
Patient 6
Patient 7
Patient 8
Patient 9
Patient 10
Patient 11
Patient 12
Change from baseline in anti-PLA2R antibodies (%)
Start of treatment
EOT
EOS
Month

EOS, end of study; EOT, end of treatment; PLA2R, phopholipase A2 receptor.

UPCR responses

- 12 patients have EOS UPCR data available; 4/12 (33%) patients did not complete 12-months of study participation per protocol, and had an earlier EOS (early EOS patients), due to adverse events (n=2) and physician's decision (n=2)
- Three of six (50%) patients from Cohort 1 and one of six (17%) patients from Cohort 2 had a UPCR decrease of >50% from baseline (**Figure 7a**)
- Most of the patients with no UPCR improvement had a shorter duration of the follow-up (early EOS)
- An assessment of serum albumin levels showed a positive change (increase) from baseline for the majority of patients at EOS (**Figure 7b**); median (range) albumin was 3.00 (2.10–4.10) g/dL at EOS
- In most patients, an early anti-PLA2R antibody titer reduction at 3 months was followed by a later UPCR response at 6 months (Pearson correlation=0.49, p=0.017) (**Figure 8**)

Figure 7. Relative change from baseline in (a) UPCR and (b) serum albumin levels at EOS

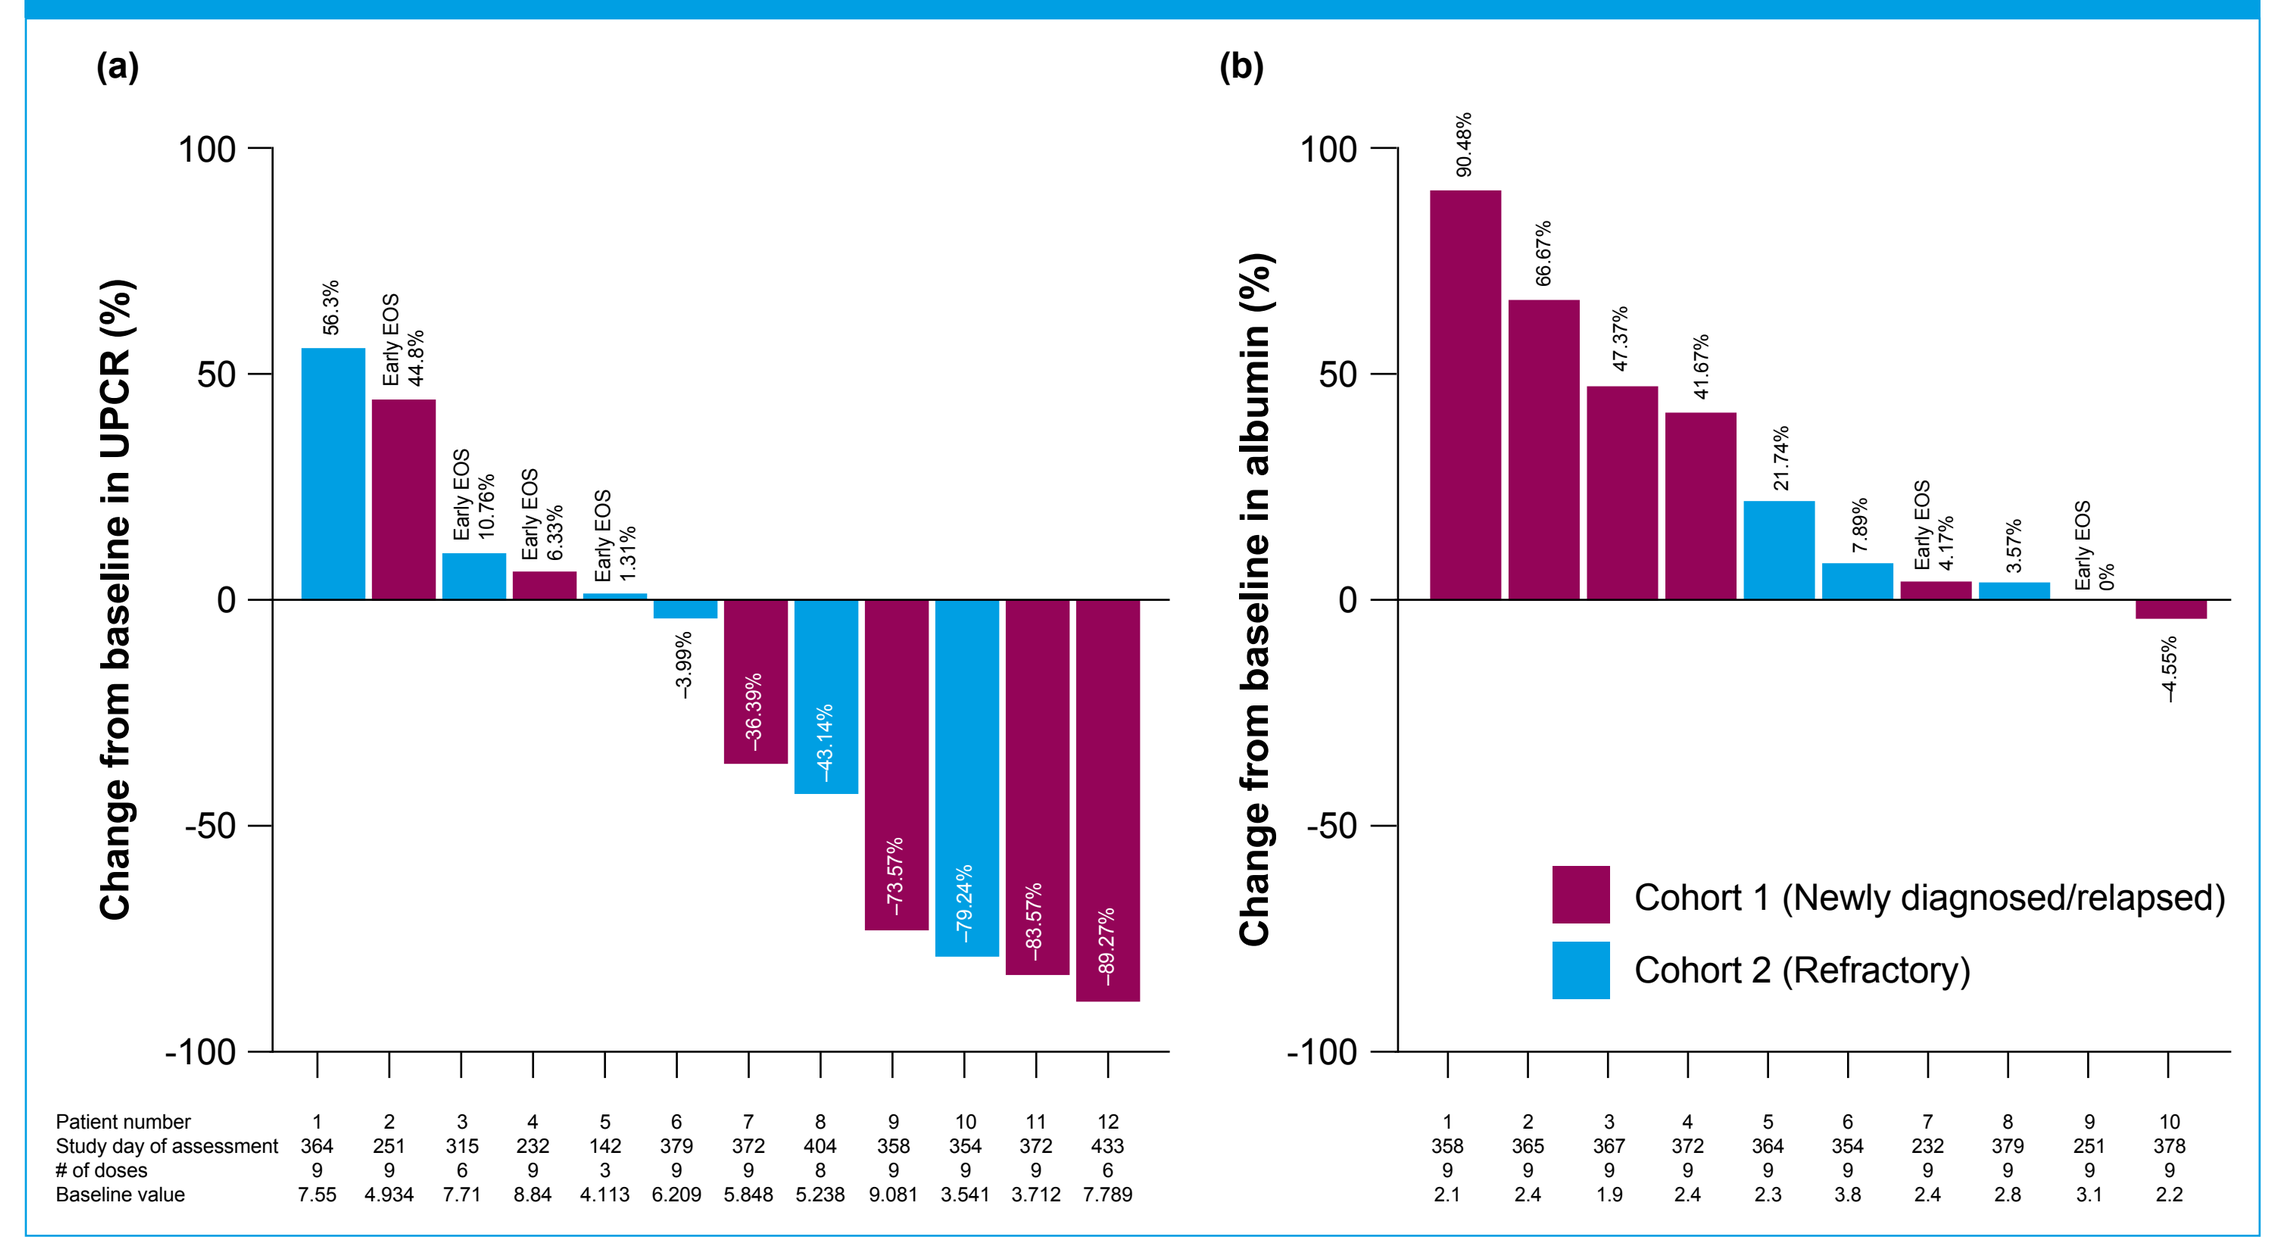
(a)
Change from baseline in UPCR (%)
56.3%
Early EOS 44.8%
Early EOS 10.76%
Early EOS 6.33%
Early EOS 1.31%
−3.99%
−36.39%
−43.14%
−73.57%
−79.24%
−83.57%
−89.27%
(b)
Change from baseline in albumin (%)
90.48%
66.67%
47.37%
41.67%
21.74%
7.89%
Early EOS 4.17%
3.57%
Early EOS 0%
−4.55%
Cohort 1 (Newly diagnosed/relapsed)
Cohort 2 (Refractory)
Patient number
Study day of assessment
of doses
Baseline value

EOS, end of study; UPCR, urine protein:creatinine ratio.

Figure 8. Correlation of anti-PLA2R antibody levels at 3 months and UPCR results at 6 months

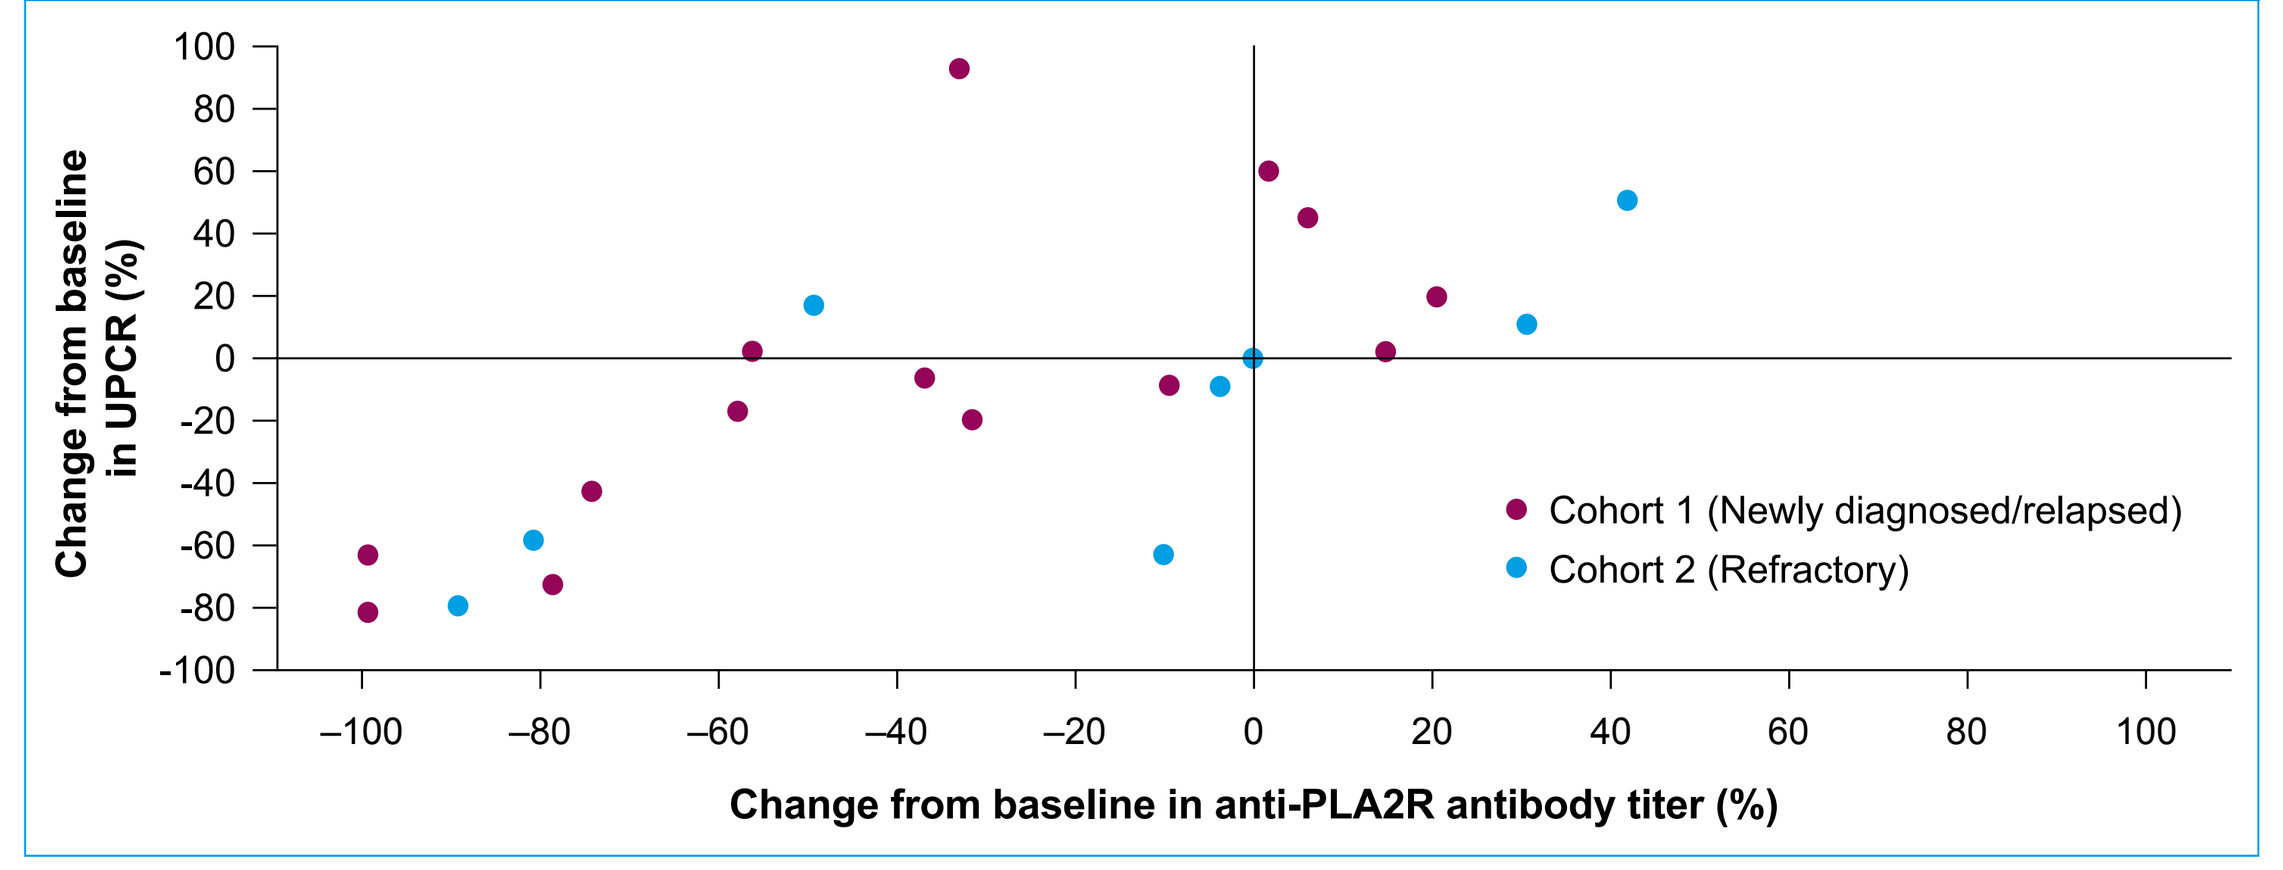
Change from baseline in UPCR (%)
Change from baseline in anti-PLA2R antibody titer (%)
Cohort 1 (Newly diagnosed/relapsed)
Cohort 2 (Refractory)

There is one missing patient on this figure due to their PLA2R % change from baseline out of the 100% range.
PLA2R, phospholipase A2 receptor; UPCR, urine protein:creatinine ratio.

Summary

- The M-PLACE proof-of-concept study is evaluating the safety and efficacy of felzartamab in patients with anti-PLA2R antibody-positive MN, including those in whom achievement of immunologic remission is considered difficult
 - Patients had medium or high anti-PLA2R antibody levels at baseline, as compared with other studies of ISTs in anti-PLA2R antibody-positive MN,[13,14] and/or patients who were refractory to previous IST
 - Increased autoantibody levels at baseline are described as a risk factor for low response rates of standard of care treatments[1,8,13,15]
- Longer follow-up of the M-PLACE study shows safety data consistent with the safety profile previously described for felzartamab[11] and its proposed mechanism of action, and as expected in this underlying population
- The early (1 week after the first felzartamab administration) and pronounced anti-PLA2R antibody responses observed in this study support its proposed mechanism of action on CD38+ plasma cells
- Independent of the treatment history of patients in the two cohorts in this study, a rapid immunologic response is followed by a clinical response in most patients, as shown by long-term UPCR responses
- In summary, this proof-of-concept study shows the potential of felzartamab to induce rapid and durable-immunologic and clinical responses in some patients with difficult-to-treat anti-PLA2R antibody-positive MN through depletion of CD38+ plasma cells

Acknowledgments

This study was sponsored by MorphoSys AG, and funded by both MorphoSys and HI-Bio. The authors would like to thank Anjali Thakur, Rainer Boxhammer, and Liliya Chamitava, MorphoSys AG, for their contribution to data analysis. Medical writing assistance was provided by Ifigeneia Mavragani, PhD, of Syneos Health, UK, and funded by MorphoSys AG.

Disclosures

About felzartamab
I-Mab and Human Immunology Biosciences, Inc., (HI-Bio) have licensed rights to develop felzartamab (MOR202/TJ202) in the greater China territory and rest of world, respectively. HI-Bio will assume development responsibility of felzartamab ex-Greater China at the end of a transition period slated for the first half of 2023. I Mab has initiated a Phase II (NCT03860038) and a Phase III (NCT03952091) study to evaluate felzartamab in patients with multiple myeloma (MM). With the productive partnership with MorphoSys, I-Mab has completed felzartamab registrational trial for third-line MM and advanced the Phase III trial for second-line MM in China.

Conflicts of interest
BHR: consulting fees from MorphoSys AG. **PMR:** consulting fees from Alexion, MorphoSys AG; research funding from Amgen. **JFMW:** advisory fees and research funding from MorphoSys AG. **SGA:** advisory fees from Nephrotic Syndrome Study Network (NEPTUNE), CureGN, Kidney Research Network, REBOOT, KDIGO, NephCure Kidney International Foundation, Karger Publishers, AstraZeneca, Aurinia, Bayer, Calliditas, ChemoCentryx, MorphoSys AG, Omeros, Travere, Zyversa. **IA:** advisory fees from ChemoCentryx, Aurinia. **PZ:** participation as PI. **SHH:** nothing to disclose. **NF:** employee of MorphoSys AG. **JC:** employee of MorphoSys US Inc. **JJ-L, SH:** employees of MorphoSys AG with stock or stock options. **BS:** consulting fees from MorphoSys AG.

References

1. Ronco P, et al. Nat Rev Dis Prim 2021;7(1):1–23.
2. Passerini P, et al. Front Immunol 2019;10:1326.
3. Bomback AS, et al. Am J Nephrol 2018;47(suppl 1):30–42.
4. Beck L, et al. N Engl J Med 2009;361(1):11–21.
5. Hofmann K, et al. Front Immunol 2018;9:835.
6. Hoxha E, et al. J Am Soc Nephrol 2014; 25(6):1357–66.
7. Bech AP, et al. Clin J Am Soc Nephrol 2014;9(8):1386–92.
8. Rovin BH, et al. Kidney Int 2021;100(4):S1–276.
9. Endell J, et al. Blood 2012;120(21):4018.
10. Boxhammer R, et al. Blood 2015;126(23):3015.
11. Rovin BH, et al. Am J Kidney Dis 2022;79(4):S3.
12. Rovin BH, et al. Kidney Int Reports 2022;7(9):2086–90.
13. Ruggenenti P, et al. J Am Soc Nephrol 2015;26(10):2545–58.
14. Scolari F, et al. J Am Soc Nephrol 2021;32(4):972–82.
15. Logt A-E van de, et al. Kidney Int 2018;93(4):1016–7.

Scan the QR code to download a PDF of this poster

Presented at the American Society of Nephrology (ASN) Annual Meeting 2022; November 3–6, 2022

*Correspondence: Prof. Brad H. Rovin, MD (Brad.Rovin@osumc.edu)